Toward LC-FAOD Genetic Diagnoses: Insights from a Rhabdomyolysis Gene Panel

Tatiele Nalin;[1*] Omar Riemersma;[1] Andrea Schenone;[1] Carla Judice;[1] Heraclio Gutierrez;[1] Daniela Visconti;[1] Omid K. Japalaghi;[1] Vanessa Rangel Miller[1]

[1]Ultragenyx Pharmaceutical Inc, Novato, CA

*TNalin@ultragenyx.com

Background

Long-chain fatty acid oxidation disorders (LC-FAOD) are rare inborn errors of metabolism leading to serious consequences due to inability to convert long-chain fat into energy.[1]

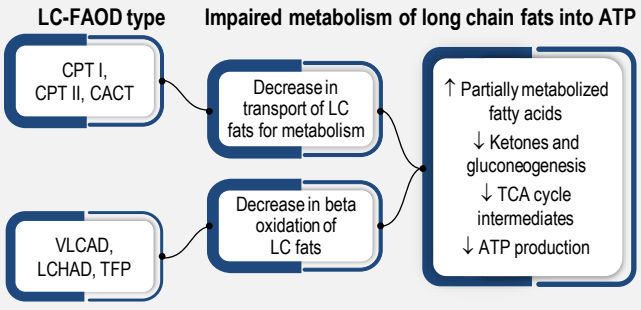
LC-FAOD type
Impaired metabolism of long chain fats into ATP
CPT I, CPT II, CACT
VLCAD, LCHAD, TFP
Decrease in transport of LC fats for metabolism
Decrease in beta oxidation of LC fats
↑ Partially metabolized fatty acids
↓ Ketones and gluconeogenesis
↓ TCA cycle intermediates
↓ ATP production

Manifestations include:

- Rhabdomyolysis, Hypotonia/Weakness, Pain, Exercise intolerance
- Cardiomyopathy, Arrhythmia
- Hypoglycemia, Steatohepatitis, Hepatomegaly, Liver failure

LC-FAOD commonly present with muscular symptoms such as muscle weakness and recurrent rhabdomyolysis

LC-FAOD nuclear genes encode mitochondrial proteins CPT I, CPT II, CACT, TFP/LCHAD, and VLCAD

LC-FAOD genes encode mitochondrial proteins necessary for energy production

Breakdown of fatty acids is necessary for energy production when sugar, or glucose, levels are low. Without this energy supply, people with LC-FAOD may have symptoms that include, but are not limited to muscle pain, muscle weakness, low blood sugar, and fatigue.[1]

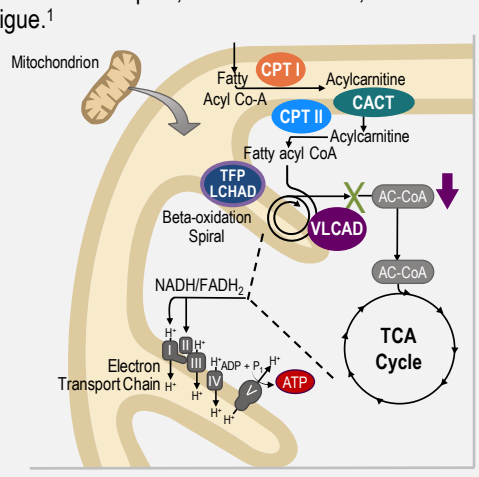
Mitochondrion
Fatty Acyl Co-A
CPT I
Acylcarnitine
CACT
CPT II
Acylcarnitine
Fatty acyl CoA
TFP LCHAD
Beta-oxidation Spiral
VLCAD
AC-CoA
AC-CoA
TCA Cycle
NADH/FADH2
Electron Transport Chain
ADP + Pi
ATP

Stylized figure based on figure 7 from reference 2.

"Fatty acid oxidation provides reducing equivalents directly to the electron transport chain and supplies substrates to the tricarboxylic acid (TCA) cycle. Oxidation of even chain fats produces only acetyl-CoA and the TCA cycle can ultimately become depleted of odd chain carbon species with excessive catabolism in patients with LC-FAODs."[2]

Program Eligibility

This program provides a no-charge, sponsored gene panel test for individuals known to have or suspected of having LC-FAOD. The patient must be in Latin America and:

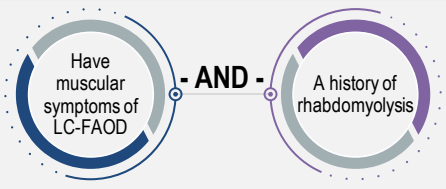
Have muscular symptoms of LC-FAOD
- AND -
A history of rhabdomyolysis

Methods

- This Program provided no-charge molecular testing, sponsored by Ultragenyx Pharmaceutical Inc.
- Individuals were tested with a comprehensive gene sequencing panel that included either six LC-FAOD genes plus 127 additional genes or four LC-FAOD genes plus 47 rhabdomyolysis-related genes.

Table 1. LC-FAOD genes

Gene	Disorder(s)
ACADVL	Very long chain Acyl-CoA dehydrogenase (VLCAD) deficiency
*CPT1A**	Carnitine palmitoyltransferase type I (CPT I) deficiency
CPT2	Carnitine palmitoyltransferase type II (CPT II) deficiency
HADHA	Long-chain 3-hydroxyacyl-CoA dehydrogenase (LCHAD) deficiency or TFP deficiency
HADHB	Trifunctional protein deficiency (TFP) deficiency
*SLC25A20**	Carnitine-acylcarnitine translocase (CACT) deficiency

**CPT1A* and *SLC25A20* were included on one of the panels. The other four LC-FAOD genes were included on both testing panels.

Variants were classified according to ACMG/AMP guidelines[3]

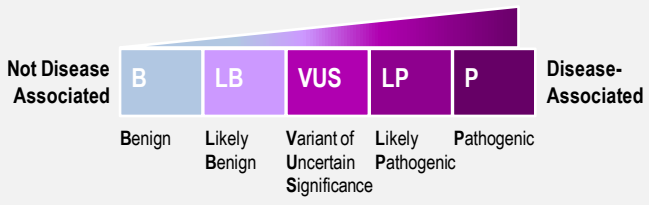
Not Disease Associated
B
LB
VUS
LP
P
Disease-Associated
Benign
Likely Benign
Variant of Uncertain Significance
Likely Pathogenic
Pathogenic

Criteria for molecular diagnoses

Positive Result:
Autosomal recessive gene = 2 Pathogenic (P)/Likely-Pathogenic (LP) variants; Autosomal dominant or X-linked gene = 1 P/LP variants

Potential Positive Result:
Autosomal recessive gene = 2 variants of which at least 1 is a Variant of Uncertain Significance (VUS); Autosomal dominant or X-linked gene = >1 variants of which at least 1 is a VUS

Results

Characteristics of Individuals Tested

- As of January 31, 2025, 166 individuals were tested from:
 - Argentina (139, 84%)
 - Brazil (13, 8%)
 - Mexico (7, 4%)
 - Chile (6, 4%)
 - Guatemala (1, <1%)

Molecular diagnoses

- Six individuals tested had a positive or potential positive genetic diagnosis for a LC-FAOD (all *CPT2*), 28 had a positive or potential positive diagnosis for other genes/disorders on the panel (**Figure 2**)
 - 7 additional individuals had variants identified but no molecular diagnosis

Figure 2. Molecular diagnoses (MDx) for *CPT2* (a) and other genes on the rhabdomyolysis panel (b and c), among 166 individuals tested

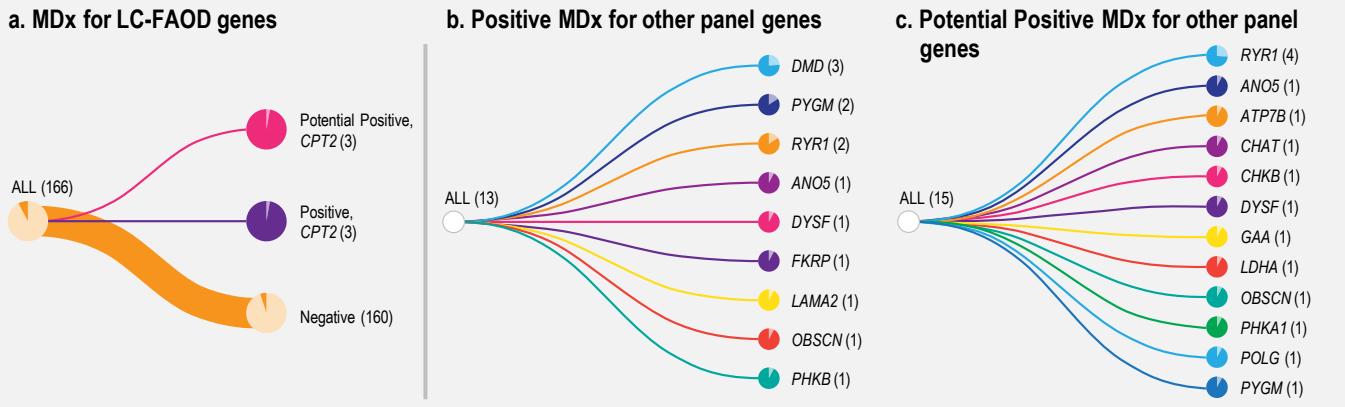
a. MDx for LC-FAOD genes
ALL (166)
Potential Positive, CPT2 (3)
Positive, CPT2 (3)
Negative (160)
b. Positive MDx for other panel genes
ALL (13)
DMD (3)
PYGM (2)
RYR1 (2)
ANO5 (1)
DYSF (1)
FKRP (1)
LAMA2 (1)
OBSCN (1)
PHKB (1)
c. Potential Positive MDx for other panel genes
ALL (15)
RYR1 (4)
ANO5 (1)
ATP7B (1)
CHAT (1)
CHKB (1)
DYSF (1)
GAA (1)
LDHA (1)
OBSCN (1)
PHKA1 (1)
POLG (1)
PYGM (1)

Table 2. Molecular diagnoses by age group

Age Group	Negative	*CPT2* Positive	*CPT2* Potential Positive	Other Gene Positive	Other Gene Potential Positive	Total
<1 year				2		**2**
1–12 years	23	1		3	2	**29**
13–20 years	32	2	2	2	2	**40**
21–40 years	4		1	4	8	**57**
>40 years	34			2	2	**38**

Figure 3. Molecular diagnostic results by country

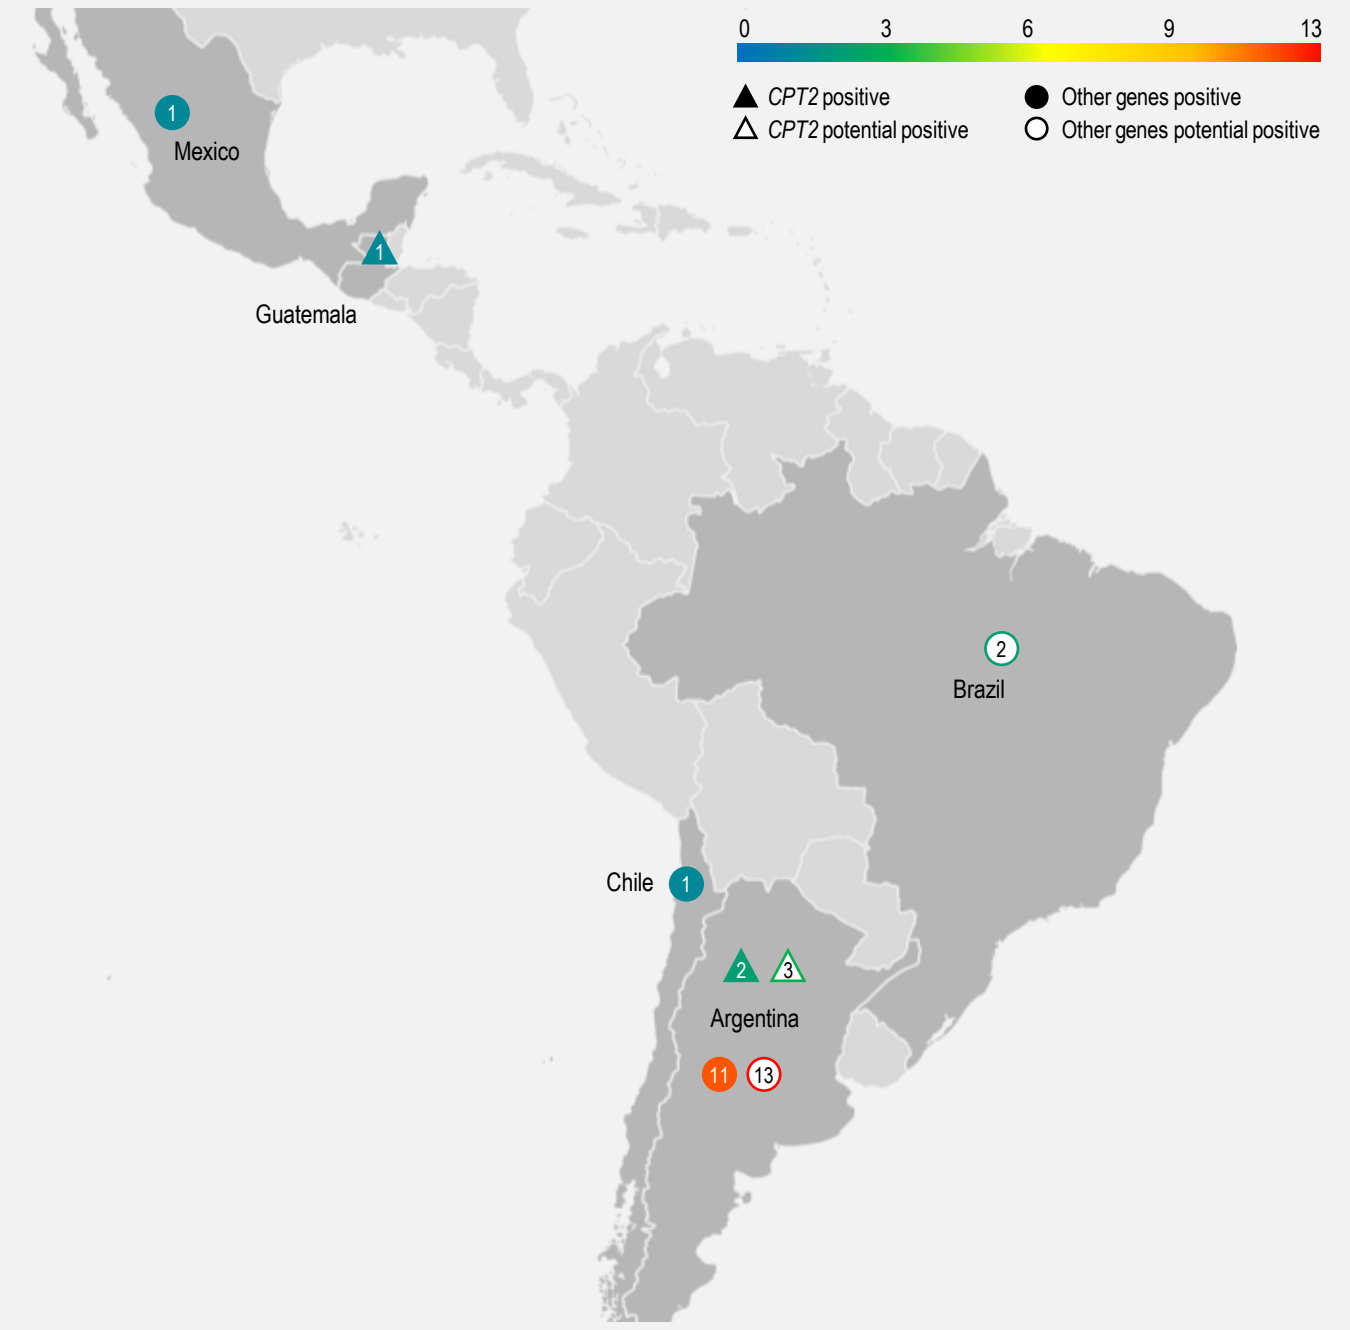
0
3
6
9
13
CPT2 positive
CPT2 potential positive
Other genes positive
Other genes potential positive
1
Mexico
1
Guatemala
2
Brazil
Chile
1
2
3
Argentina
11
13

CPT2 variants

- 12 *CPT2* variants (6 unique) were identified among 6 individuals
 - 4P, 2 VUS

Table 3. *CPT2* variants and characteristics

Variant (cDNA)	Amino Acid Change	Frequency	Classification	Variant type	Effect Type
c.110_111dup*	p.Ser38Alafs*36	2	Pathogenic	Duplication	Frameshift
c.1511C>T*	p.Pro504Leu	1	Pathogenic	SNV	Missense
c.338C>T*	p.Ser113Leu	5	Pathogenic	SNV	Missense
c.340G>C	p.Ser113Leu	1	VUS	SNV	Missense
c.534_558delinsT*	p.Leu178_Ile186 delinsPhe	1	Pathogenic	Deletion/insertion	Splice acceptor/ donor
c.587C>T*	p.Pro196Leu	2	VUS	SNV	Missense

*Additional data regarding these variants are available on the rare disease genes database website.[4] VUS = variant of uncertain significance; SNV = single nucleotide variant.

Acknowledgments and Disclosures

We recognize and thank the individuals tested through this program.

All authors are employees and shareholders of Ultragenyx Pharmaceutical Inc.

Third-party writing support was provided by Kariena Dill and paid for by Ultragenyx Pharmaceutical Inc.

Graphic design support was provided by Allison Saviano of Sephirus Communications, Inc., and paid for by Ultragenyx Pharmaceutical Inc.

References

1. www.faodinfocus.com
2. Vockley, et al. *Mol Genet Metab.* 2015; v116.
3. Nykamp K, et al. *Genet Med.* 2017 Oct; 19(10).
4. https://www.rarediseasegenes.com/lc-faod

Conclusions

- This study highlights the importance of genetic testing with a comprehensive gene panel for diagnosing LC-FAOD in patients with muscular symptoms and/or rhabdomyolysis.
- In addition to LC-FAOD, a diverse genetic landscape of myopathies were diagnosed across ages, suggesting gaps in a timely and accurate diagnosis.
- These findings also highlight the value of expanding newborn screening programs to include LC-FAOD and other treatable disorders, facilitating earlier intervention and improved long-term outcomes.